P1517

GLATIRAMER ACETATE SLOWS DISABILITY PROGRESSION – RESULTS FROM A 6-YEAR ANALYSIS OF THE UK RISK SHARING SCHEME

Gavin Giovannoni[1,2], Peter Brex[3], Montu Sumra[4], Ewan Walters[4], Klaus Schmierer[1,2]

[1] Blizard Institute (Neuroscience), Queen Mary University of London, London, UK
[2] Neuroscience Clinical Academic Group, The Royal London Hospital (Barts Health NHS Trust), London, UK
[3] Department of Neurology, Kings College Hospital, London, UK
[4] Medical Department, Teva UK Limited, Castleford, UK

ECTRIMS · 7th-10th October 2015 · Barcelona, Spain

INTRODUCTION

- The UK Multiple Sclerosis Risk Sharing Scheme (RSS) was established in 2002 following a National Institute for Health and Care Excellence (NICE) appraisal of the disease-modifying therapies (DMTs) glatiramer acetate (GA) and the beta interferons[1,2]
- The RSS provided eligible UK multiple sclerosis (MS) patients with access to these DMTs, and in the form of a 10-year observational study, monitored a cohort of them for long-term clinical and cost-effectiveness[1]
- The aggregate results of the 6-year data from this study were published recently, and demonstrated that, as a group, the RSS drugs were both clinically and cost effective when modelled over 20 years (Figure 1)[3]

Figure 1: Progression and utility ratios for 6-year RSS data in aggregate[3]

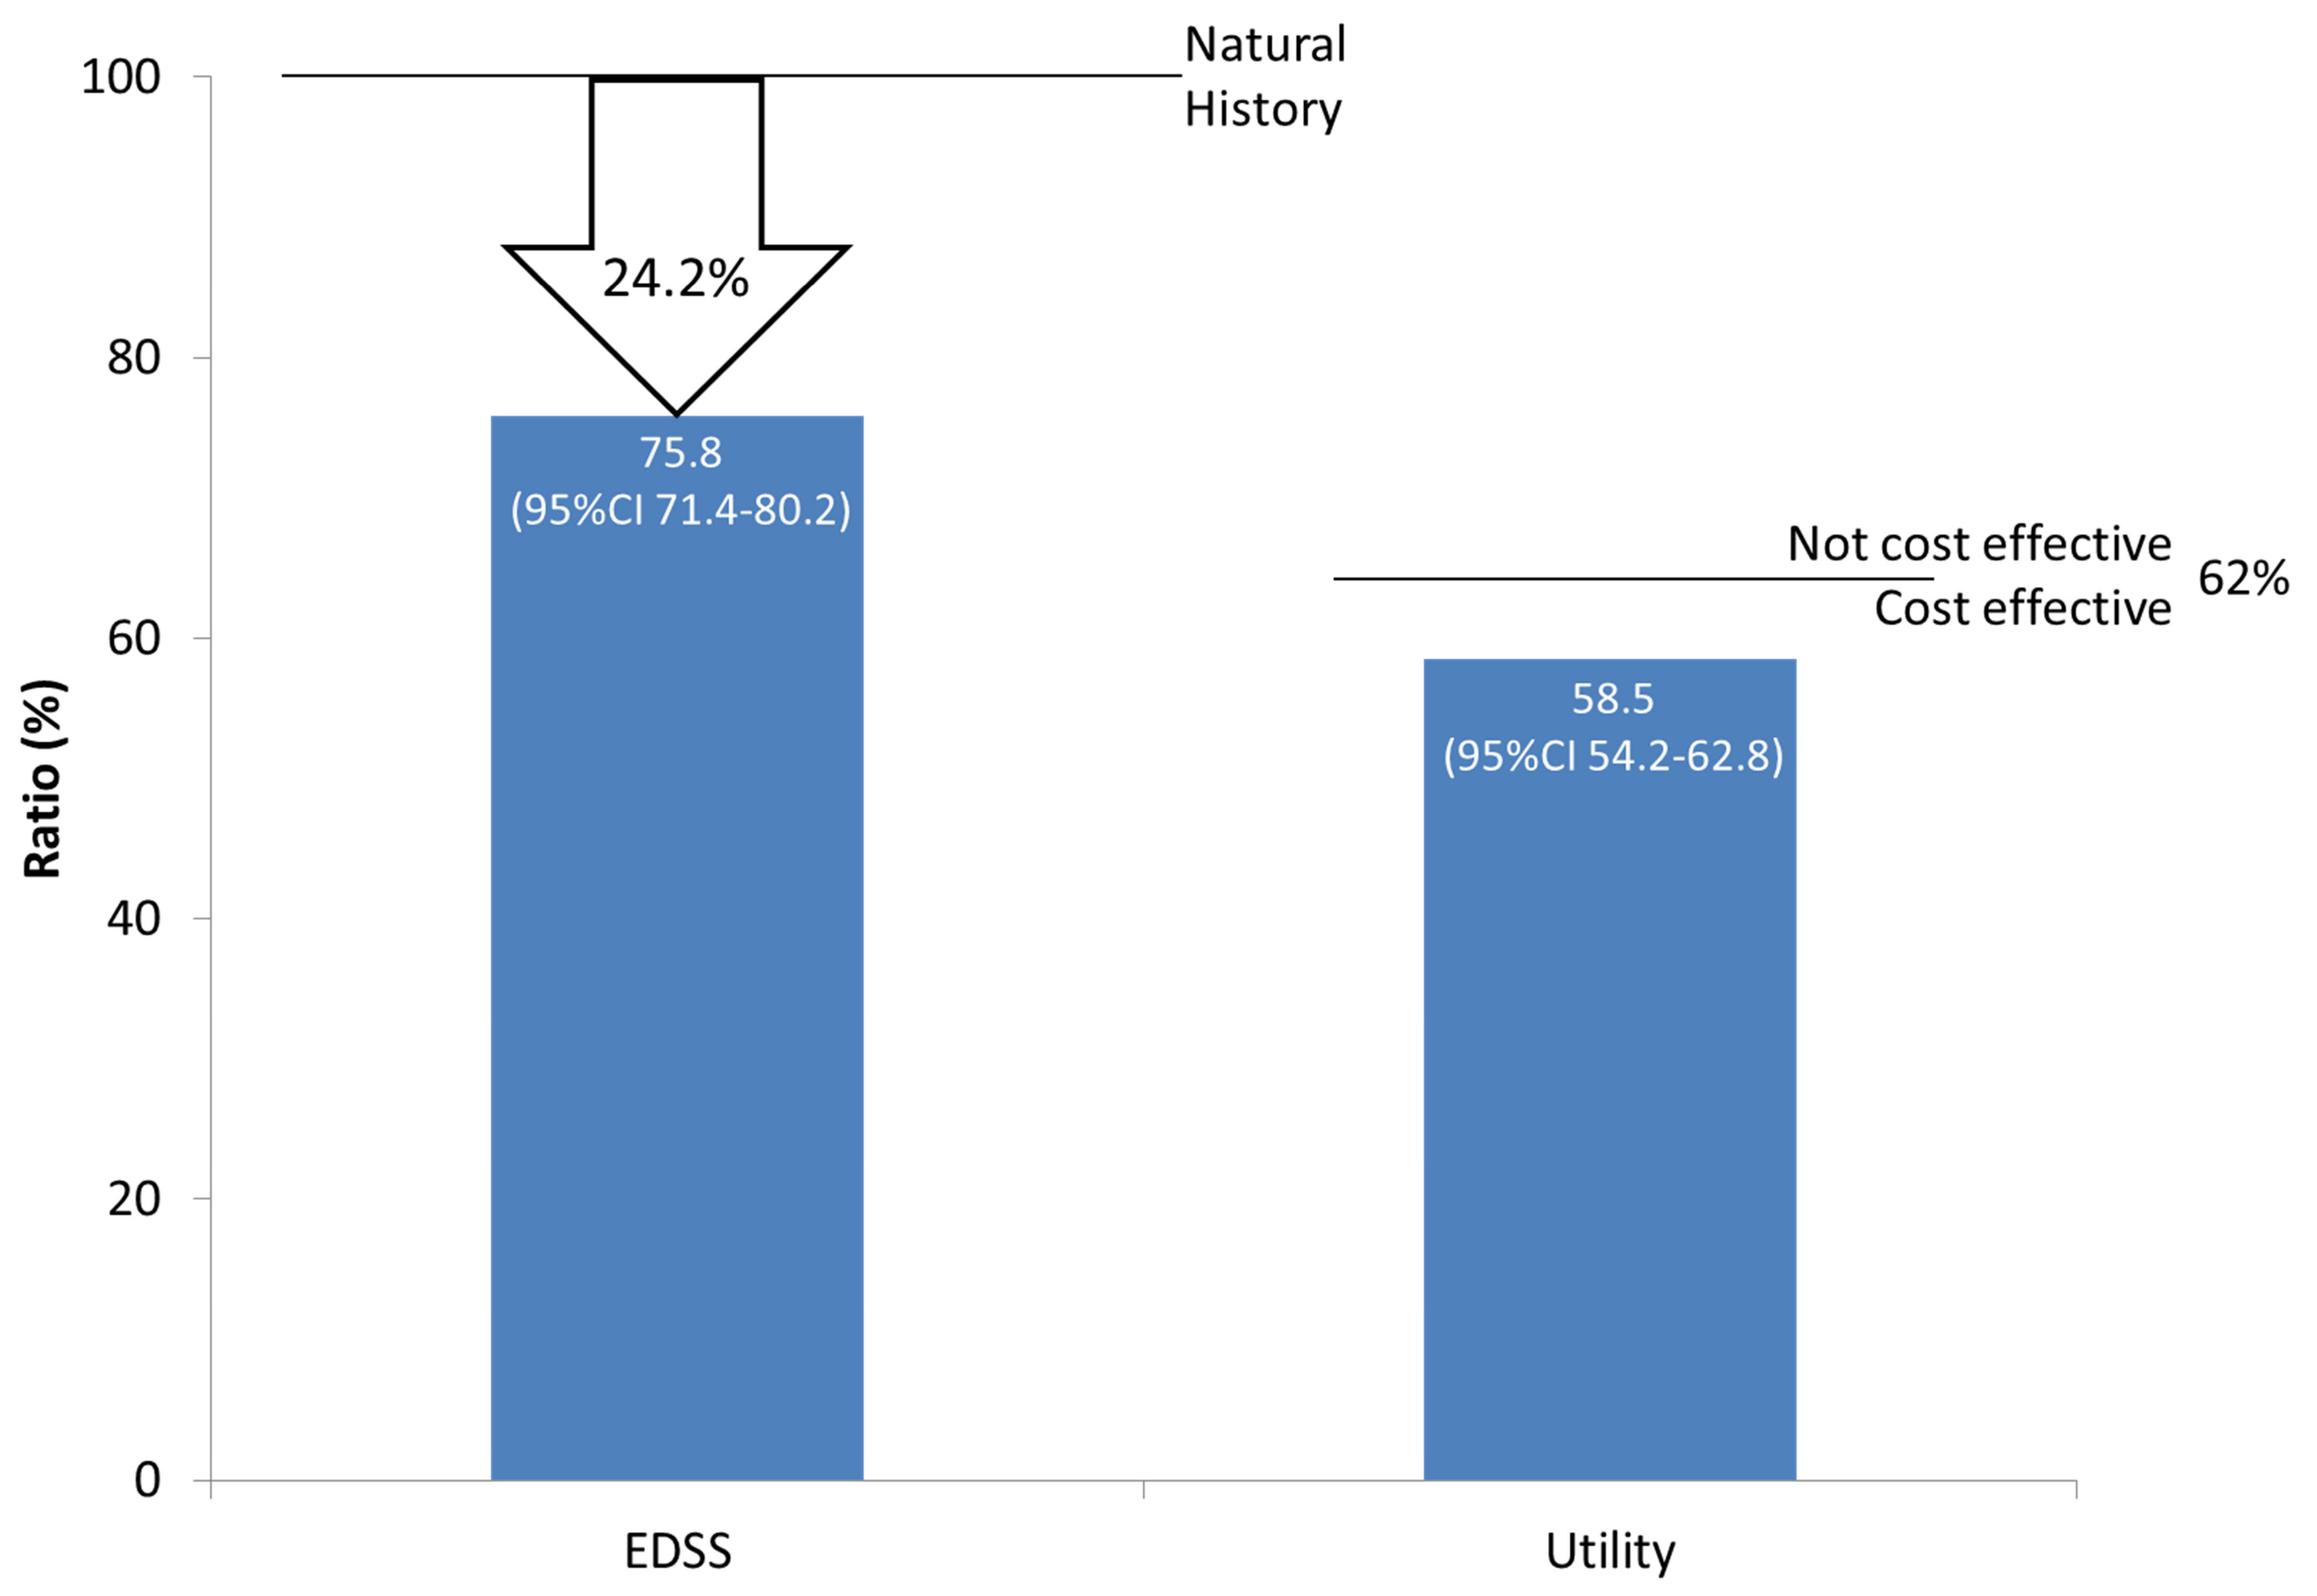
Natural History
24.2%
75.8
(95%CI 71.4-80.2)
Not cost effective
Cost effective
62%
58.5
(95%CI 54.2-62.8)
Ratio (%)
100
80
60
40
20
0
EDSS
Utility

Data derived from continuous Markov model. A ratio of less than 100% for EDSS implies slower than expected progression on DMT treatment compared to the untreated cohort. A utility progression ratio of 62% or lower means that utility progression for the drugs in aggregate is in line with, or slower than, the NICE target

PURPOSE

- To report the RSS 6-year clinical and cost effectiveness data for GA

METHODS

- Patients with relapsing MS fulfilling the Association of British Neurologists (ABN) 2001 criteria for treatment with DMTs were enrolled in the RSS, with treatment choice subject to patient and physician preference
- To demonstrate cost effectiveness, GA, in line with the other drugs under the RSS, was assigned an individual target reduction in Expanded Disability Status Scale (EDSS) worsening, predicated on data from its registration studies
- A pre-specified deviation of 10% about this target was set, above or below which the price of GA would decrease or increase, respectively
- The comparator cohort for the RSS, a "virtual placebo group", was modelled using data from the British Columbia Multiple Sclerosis database (BCMS) who met the same eligibility criteria (data collection 1980-1996)
- Change in EDSS and quality of life (QoL) was compared between patients on GA and the untreated comparator group using a continuous Markov model over a 20-year time horizon
- Primary outcomes were the progression ratio (treated *vs* untreated) measured in both EDSS score and utility
- A ratio of less than 100% for EDSS implied slower than expected progression on treatment with GA compared to the untreated cohort

RESULTS

- 978 patients starting GA were enrolled in the study, with an average age of 30.0 years at onset of disease (Table 1)
- The baseline characteristics of the two cohorts were broadly similar, although the GA cohort was older at symptom onset and had a higher EDSS at baseline than the BCMS cohort

Table 1: Baseline characteristics of GA patients in RSS cohort and BCMS database

	GA cohort (n=978)	BCMS (n=898)
Sex		
- Men	223 (23%)	232 (26%)
- Women	755 (77%)	666 (74%)
Age at onset (years)		
- Mean	30.0 (8.05)	29.2 (8.7)
- Median	30 (24-35)	28 (23-35)
EDSS at baseline		
- Mean	3.07 (1.53)	2.44 (1.70)
- Median†	3 (2-4)	2 (1-3.5)
Mean follow-up	5.23 (1.33)	6.4 (3.5)

Data are n (%), mean (SD), or median (IQR). † EDSS scores are half-integers

- GA led to a 30.3% reduction in EDSS progression (ratio 69.7) and 55.8% (ratio 44.2) reduction in QoL worsening (Figure 2)
- In absolute terms, patients who received GA had a mean EDSS score that was 0·404 units less than would have been predicted off therapy

Figure 2: Progression and utility ratios for GA using 6-year RSS data

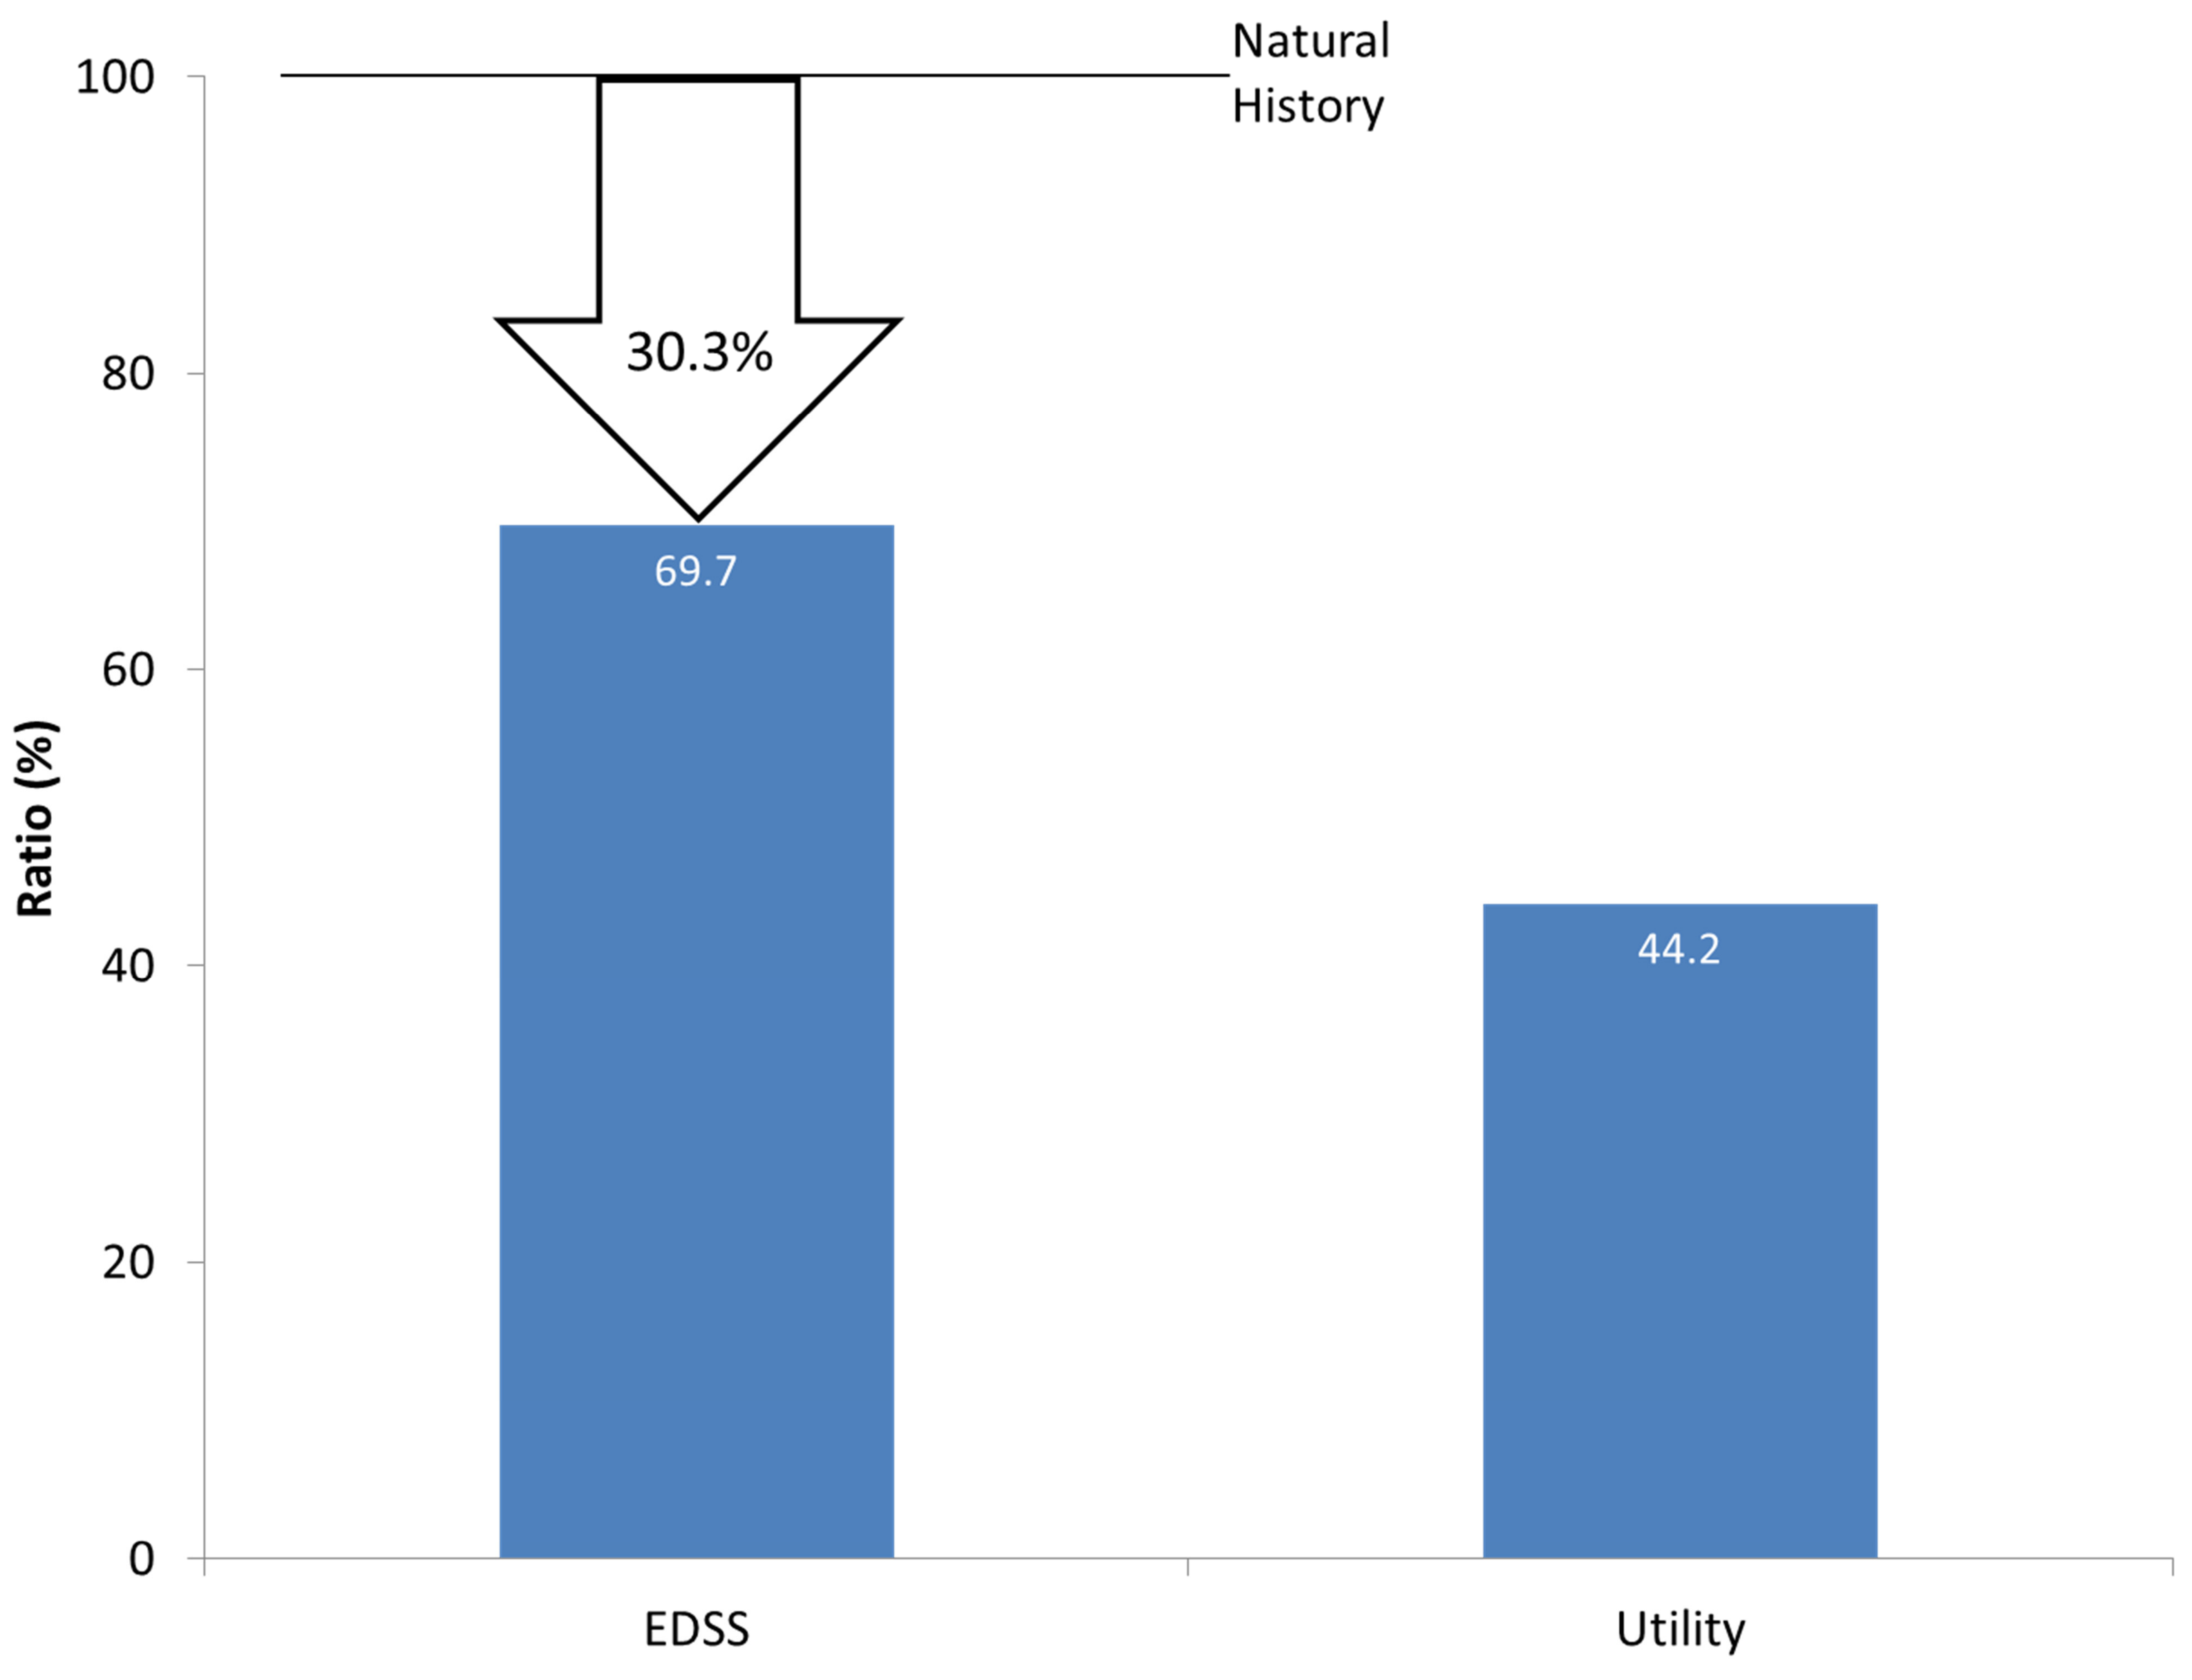
Natural History
30.3%
69.7
44.2
Ratio (%)
100
80
60
40
20
0
EDSS
Utility

- The beneficial effects of GA were significantly (>10%) above its pre-determined cost-effectiveness target enabling a price increase as agreed at the outset of the RSS

CONCLUSIONS

- This 6-year analysis supports the long-term efficacy of GA in terms of disability and QoL in patients with relapsing MS
- The cost effectiveness of GA was greater than predicted at the outset of the RSS, when modelled using the 6-year data over a 20-year time horizon
- Forthcoming analyses of the 8- and 10-year RSS data will provide further insight into the long-term benefits of GA

REFERENCES

1. Department of Health. HSC 2002/004; Feb 2002. 2. NICE technology appraisal TA32; Jan 2002.
3. Palace J *et al*. *Lancet Neurol* 2015; 14: 497-505.

ACKNOWLEDGEMENTS

- The authors would like to thank Dr Jacqueline Palace (Oxford University Hospitals Trust, Oxford, UK) and Dr Martin Duddy (Newcastle upon Tyne Hospitals, Newcastle upon Tyne, UK) for reviewing the abstract prior to submission and Dr John Fullarton (Strategen Ltd, Basingstoke, UK) who undertook the analyses of the GA data

DISCLOSURES

- Source of funding: Health Departments of England, Wales, Scotland and Northern Ireland, Teva Pharmaceuticals Industries, Biogen Idec, Merck Serono, Bayer Schering Pharmaceuticals, UK National Institute of Health Research's Health Technology Assessment Programme
- **Gavin Giovannoni** has received compensation for serving as a consultant or speaker for, or has received research support from AbbVie, Bayer Schering, Biogen, Canbex, Eisai, Elan, Five Prime Therapeutics, Genzyme, Genentech, GlaxoSmithKline, Ironwood Pharmaceuticals, Merck Serono, Novartis, Pfizer, Roche, Sanofi-Aventis, Synthon BV, Teva, UCB and Vertex Pharmaceuticals. **Peter Brex** has received honoraria from Teva, Genzyme, Biogen, Merck Serono and Roche for attending advisory boards or speaking at meetings. He has attended meetings as the guest of Biogen, Merck Serono and Genzyme. His department has received a grant from Biogen to support service developments. **Montu Sumra** is an employee of Teva UK Limited. **Ewan Walters** is an employee of Teva UK Limited. **Klaus Schmierer** is a PI on trials sponsored by Novartis, Roche and Teva. He has received honoraria from Novartis, Biogen, Teva, Merck Serono and Merck Inc for attending advisory boards or speaking at meetings, and has received travel support from Genzyme

UK
Teva UK Limited
Teva UK Limited
Ridings Point, Whistler Drive, Castleford, WF10 5HX
T: 01977 628500 F: 01977 628799
www.tevauk.com

UK/UKCPX/15/0081
Date of Preparation: September 2015